Sabirnetug (ACU193) Lowers CSF Levels of Synaptic Biomarkers in INTERCEPT-AD Phase 1 Study in Early AD

Erika N Cline,[1] Elizabeth Johnson,[1] Karen Sundell,[1] Daniel Antwi-Berko,[2] Marleen JA Koel-Simmelink,[2] Charlotte Teunissen,[2] Eric Siemers,[1] Hao Zhang,[1] Maddelyn Hyland,[1] Gopalan Sethuraman,[1] Hugo Vanderstichele,[1,3] June Kaplow,[1] Robert A Dean,[1,4] Jasna Jerecic[1]

[1]Acumen Pharmaceuticals, Inc. (USA), [2]Department of Laboratory Medicine, Neurochemistry Lab, Amsterdam UMC (the Netherlands), [3]Biomarkable (Belgium), [4]Department of Pathology & Laboratory Medicine, Indiana University School of Medicine (USA)

Introduction

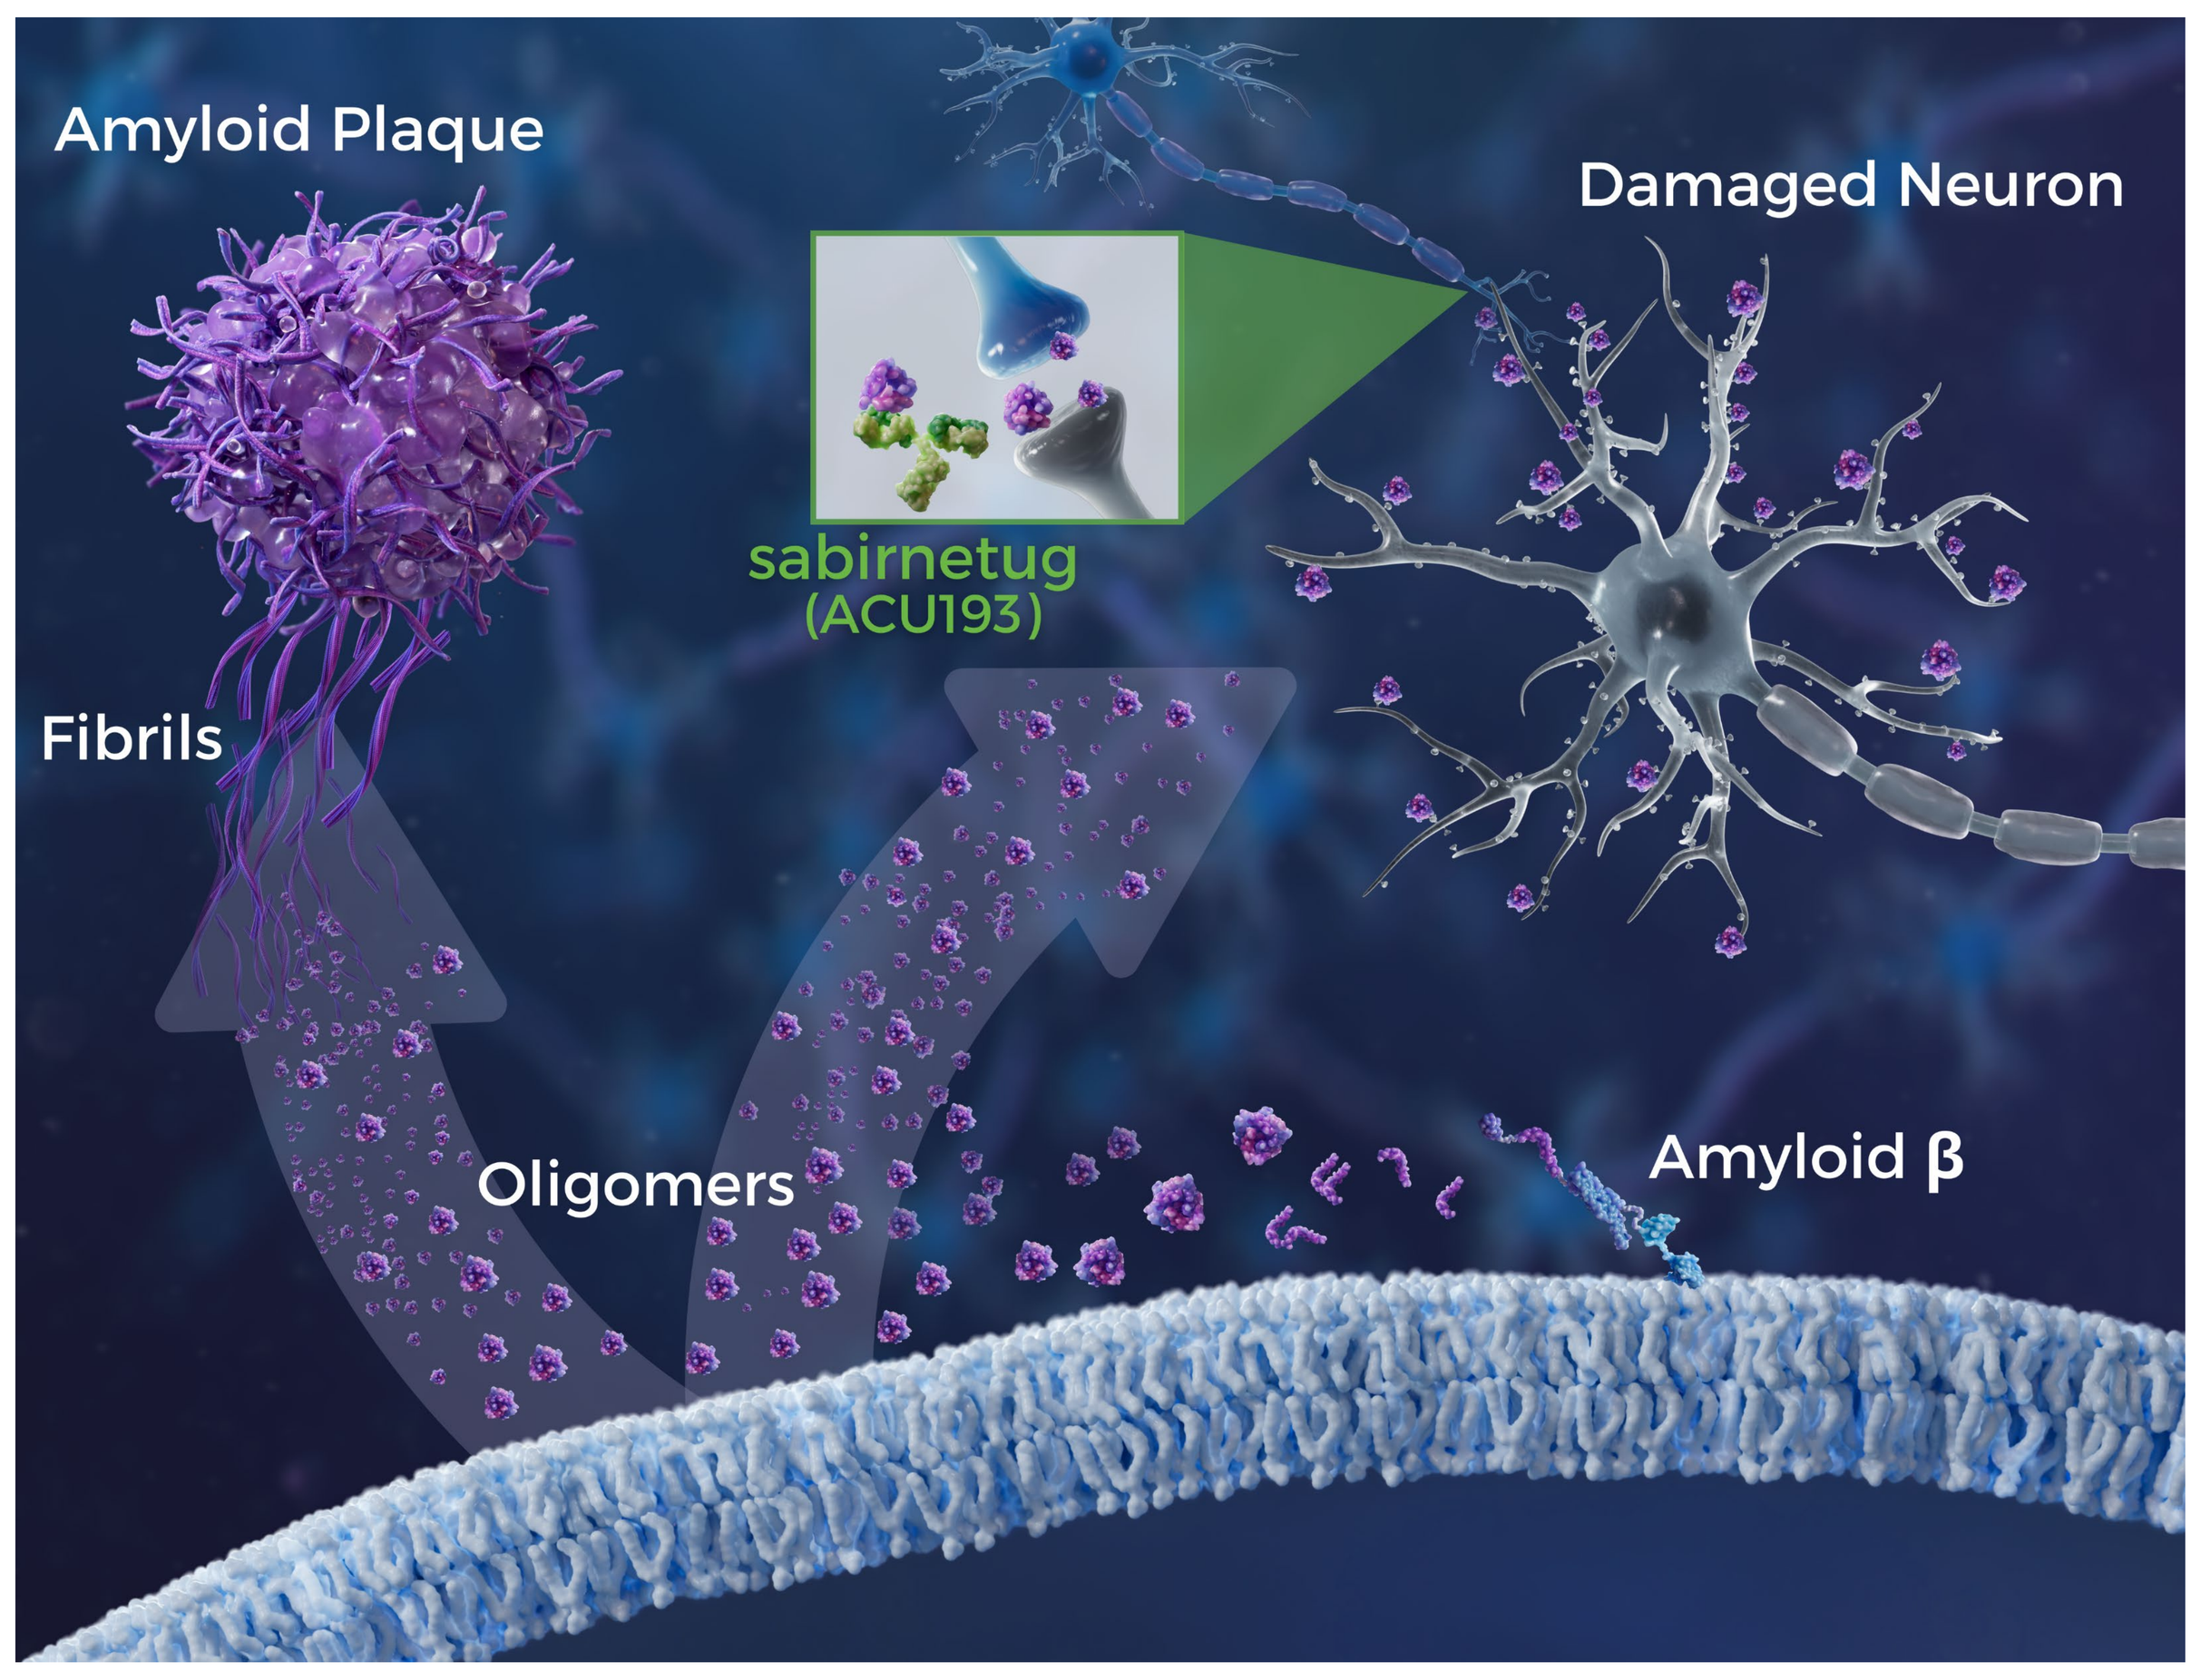

Amyloid Plaque
Damaged Neuron
sabirnetug
(ACU193)
Fibrils
Oligomers
Amyloid β

Figure 1. Sabirnetug (ACU193) is highly selective for soluble, synaptotoxic amyloid β oligomers (AβOs). In human neuronal membranes, Aβ monomers are cleaved from the amyloid precursor protein. Due to high hydrophobicity, Aβ monomers readily aggregate to oligomers (including protofibrils) or to larger insoluble amyloid fibrils, the primary component of amyloid plaques. The solubility of AβOs enables them to diffuse in the cellular milieu and bind to neuronal synapses. Their synaptic toxicity elicits downstream effects such as tau hyperphosphorylation, calcium dysregulation, and inhibition of long-term potentiation, ultimately resulting in the neuronal degeneration and cognitive impairment associated with Alzheimer's disease (AD).[1] **Sabirnetug is > 500-fold selective for AβOs over Aβ monomers[2] and > 85-fold selective for AβOs over Aβ fibrils.[3]**

Sabirnetug's proposed mechanism of action is to block Aβ oligomer binding to neuronal synapses

Methods

INTERCEPT-AD Trial Design

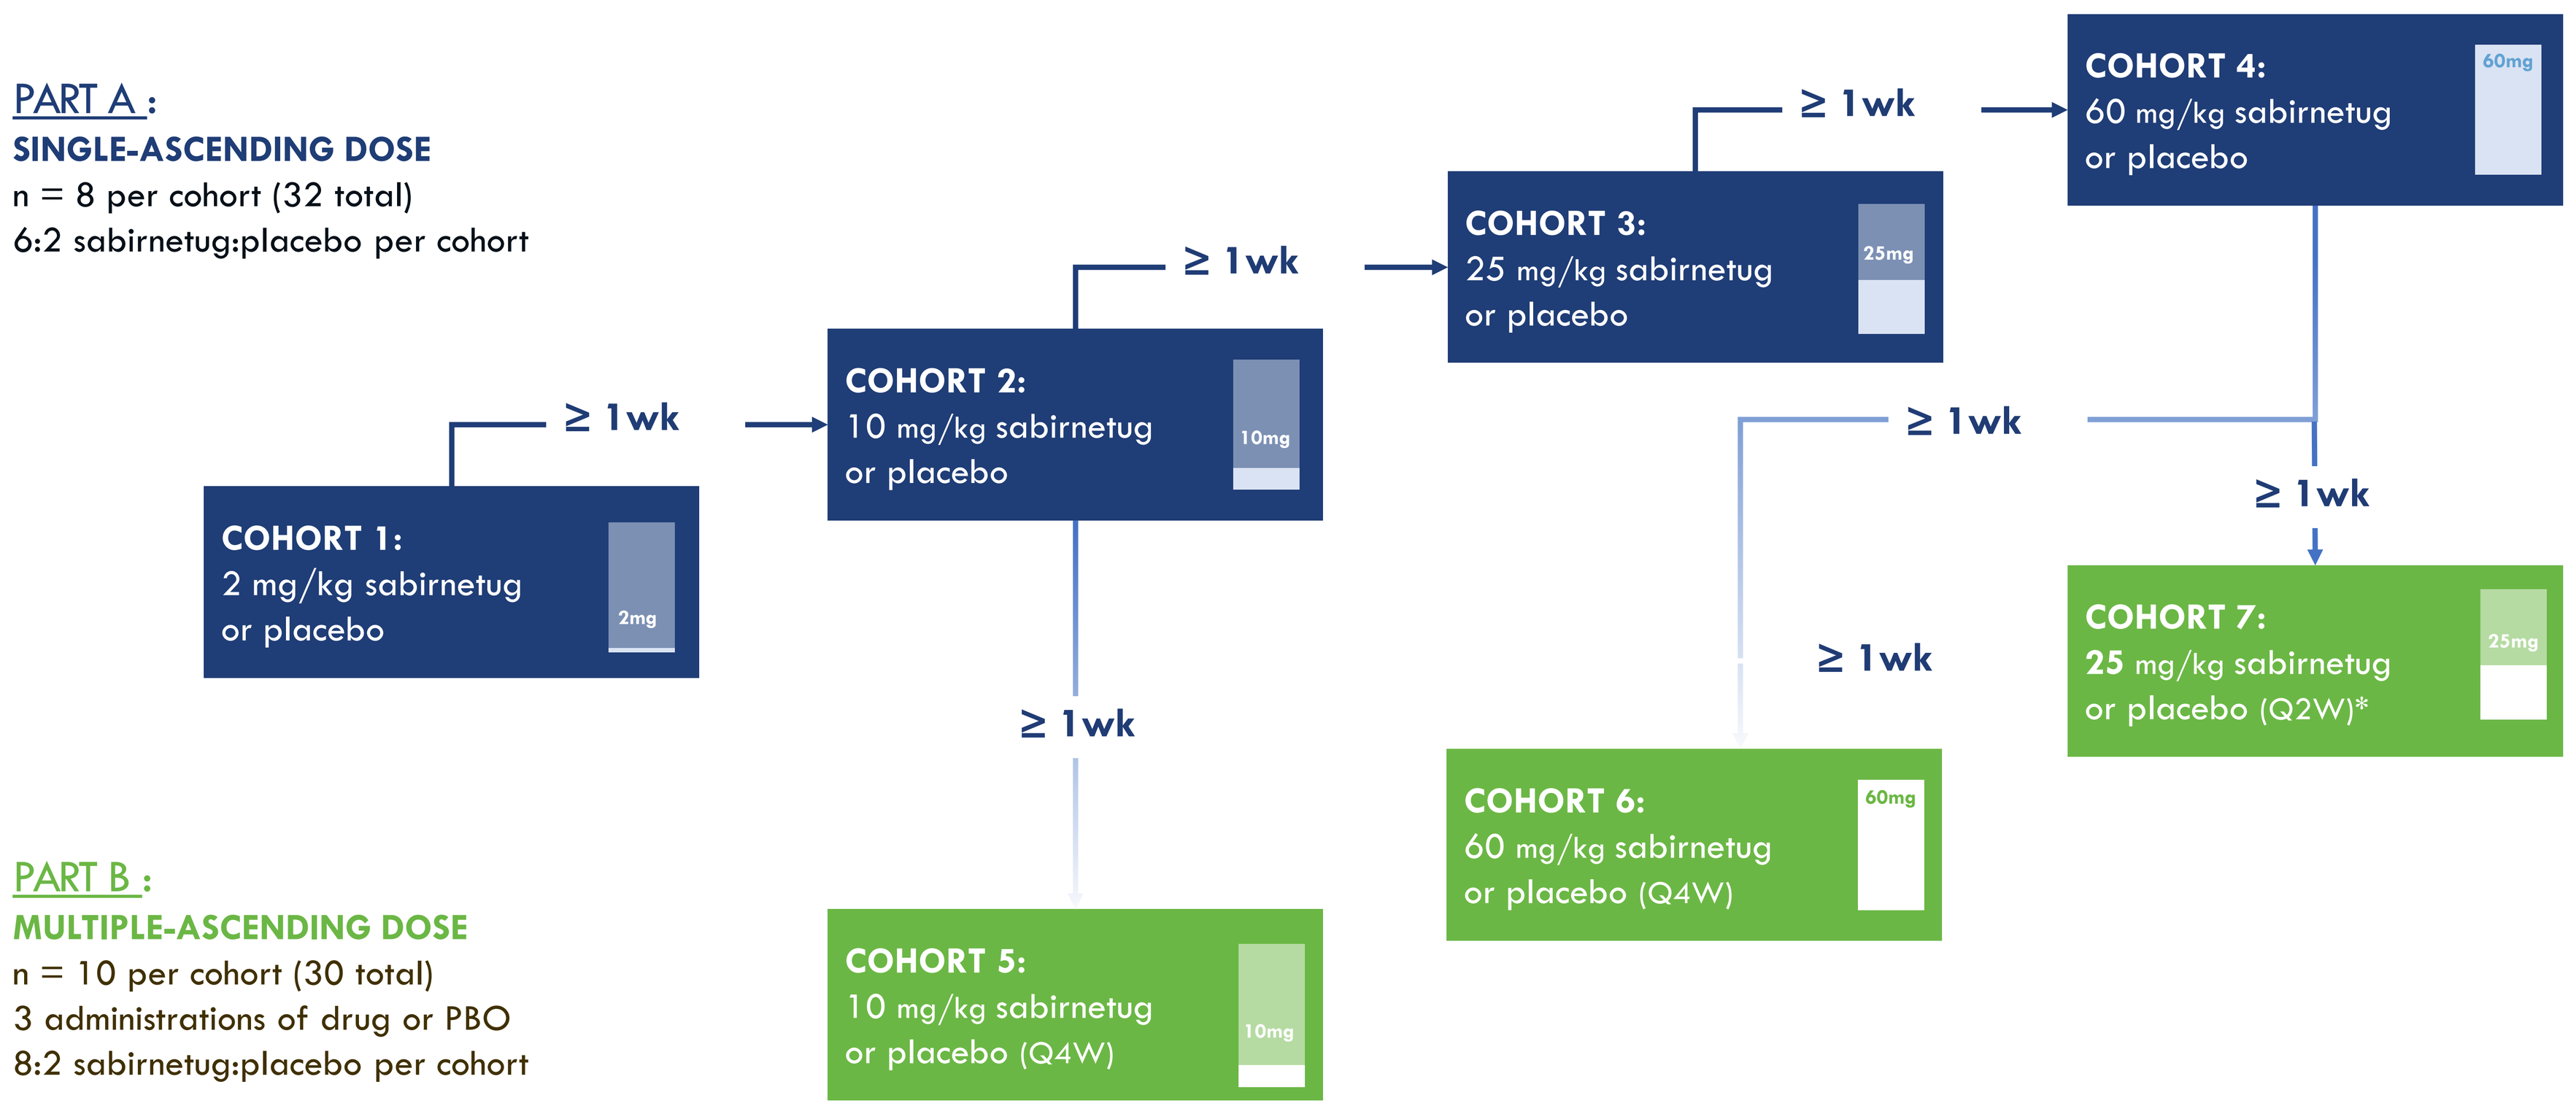

PART A :
SINGLE-ASCENDING DOSE
n = 8 per cohort (32 total)
6:2 sabirnetug:placebo per cohort
COHORT 1:
2 mg/kg sabirnetug
or placebo
2mg
≥ 1wk
COHORT 2:
10 mg/kg sabirnetug
or placebo
10mg
≥ 1wk
COHORT 3:
25 mg/kg sabirnetug
or placebo
25mg
≥ 1wk
COHORT 4:
60 mg/kg sabirnetug
or placebo
60mg
≥ 1wk
≥ 1wk
≥ 1wk
≥ 1wk
PART B :
MULTIPLE-ASCENDING DOSE
n = 10 per cohort (30 total)
3 administrations of drug or PBO
8:2 sabirnetug:placebo per cohort
COHORT 5:
10 mg/kg sabirnetug
or placebo (Q4W)
10mg
COHORT 6:
60 mg/kg sabirnetug
or placebo (Q4W)
60mg
COHORT 7:
25 mg/kg sabirnetug
or placebo (Q2W)*
25mg

Figure 2. INTERCEPT-AD was a Phase 1 clinical trial testing the safety, pharmacokinetics, and pharmacodynamics **of sabirnetug (ACU193) in mild cognitive impairment (MCI) and mild dementia due to AD** (NCT04931459). The study was conducted in two parts: A) single-ascending dose (top, navy) & B) multiple-ascending dose (bottom, green). The dosing regimen and sample sizes for each of the 7 cohorts are shown in the schematic. **CSF** was drawn from each study participant at two timepoints: (1) before the first dose; and (2) 7-21 days after the last dose. **EDTA-plasma** was collected at four timepoints in the SAD cohorts: 0, 0.5, 6, & 10 weeks after first dose. Three timepoints were collected for MAD cohorts: (1) before the first dose; (2) 1, 2, or 6 weeks after first dose in Cohorts 5, 6, 7, respectively; and (3) 10, 20, or 10 weeks after first dose in Cohorts 5, 6, 7, respectively. *Q2W = every 2 weeks; Q4W = every 4 weeks.*

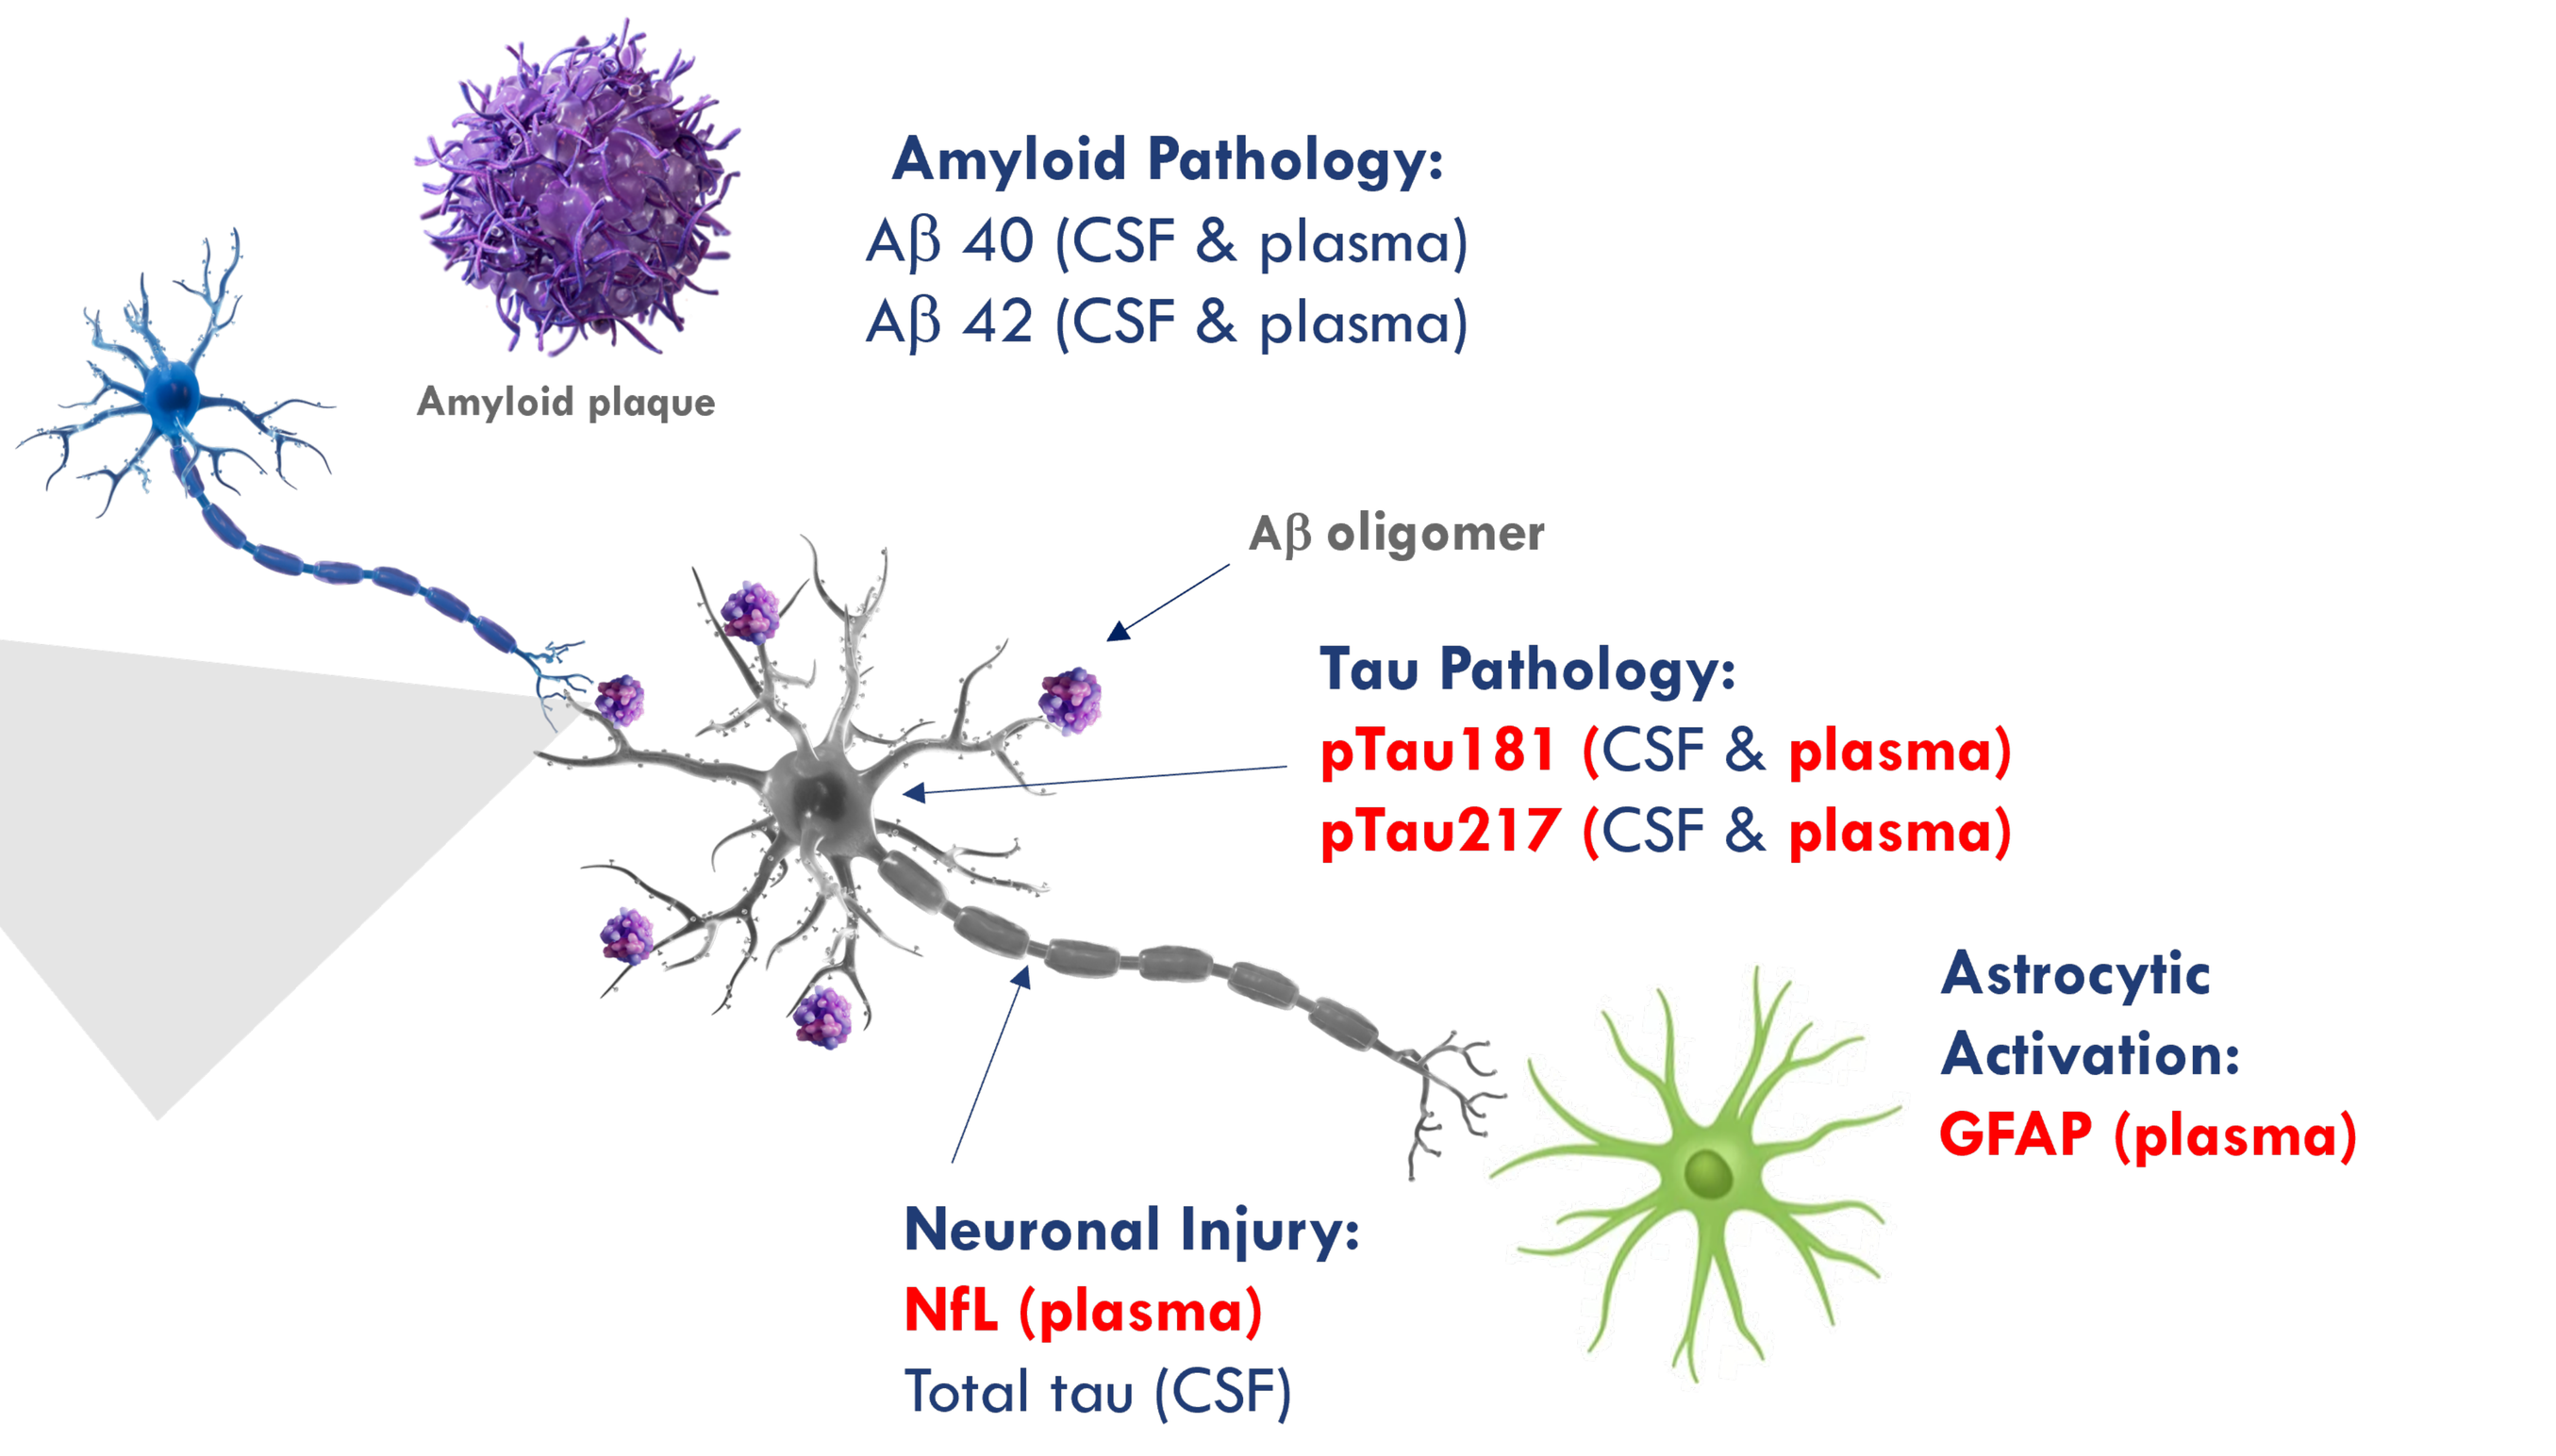

Amyloid plaque
Amyloid Pathology:
Aβ 40 (CSF & plasma)
Aβ 42 (CSF & plasma)
Aβ oligomer
Tau Pathology:
pTau181 (CSF & plasma)
pTau217 (CSF & plasma)
Astrocytic
Activation:
GFAP (plasma)
Neuronal Injury:
NfL (plasma)
Total tau (CSF)

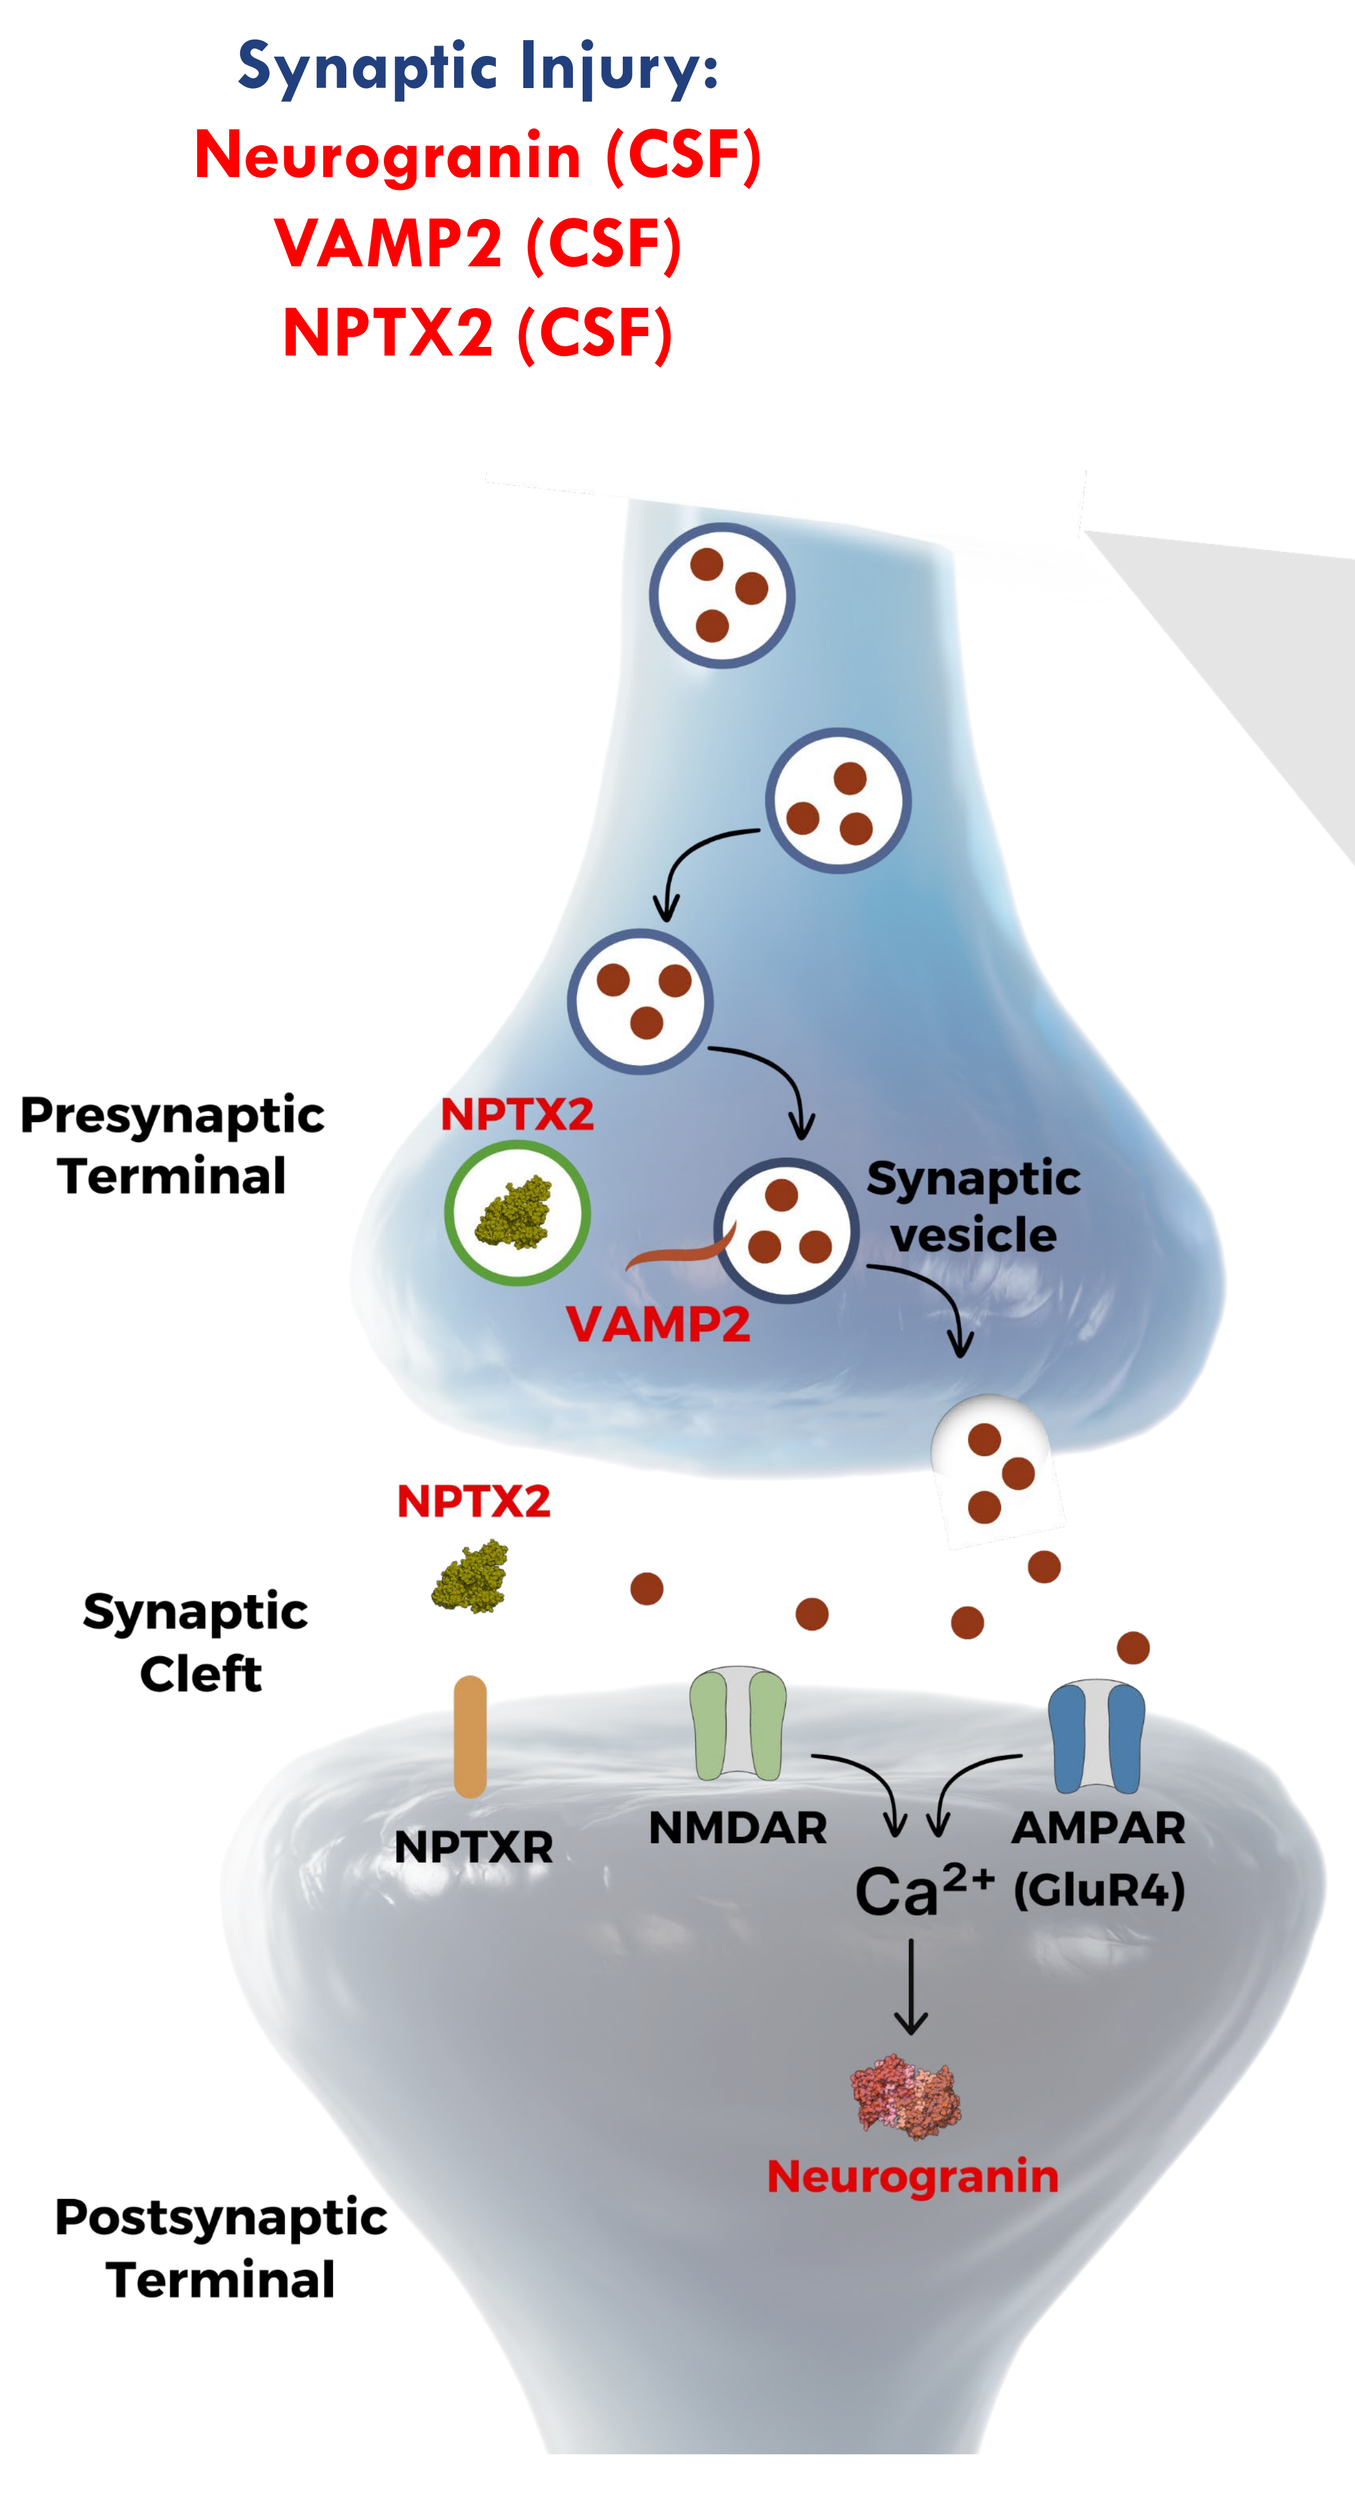

Synaptic Injury:
Neurogranin (CSF)
VAMP2 (CSF)
NPTX2 (CSF)
Presynaptic
Terminal
NPTX2
Synaptic
vesicle
VAMP2
NPTX2
Synaptic
Cleft
NPTXR
NMDAR
AMPAR
Ca2+ (GluR4)
Neurogranin
Postsynaptic
Terminal

Key ATX(N) & Synpatic Biomarkers Measured in CSF & Plasma

Figure 3. In INTERCEPT-AD, multiple ATX(N) biomarkers (A = Aβ pathway, T = tau-mediated pathophysiology, X = additional pathophysiological mechanisms such as synaptic dysfunction, N = neurodegeneration[4]) were measured in CSF and EDTA-plasma, as indicated in the graphic above. **Biomarkers for which data are presented in this poster are highlighted in red.** Results for Aβ 42/40 & the tau proteoforms in CSF were presented at the 2024 AD/PD™ meeting in Lisbon, Portugal.

Left inset) A synapse is enlarged to show the neuronal localization of the three synaptic biomarkers measured in CSF: neurogranin (P75 truncated form measured), vesicle associated membrane protein 2 (VAMP2), and neuronal pentraxin 2 (NPTX2). Neurogranin is a post-synaptic, calcium regulating protein located in dendritic spines that is involved in long-term potentiation & depression.[5] VAMP2 is a component of synaptic vesicles, functioning in neurotransmitter release and the post-synaptic vesicle trafficking of glutamate receptor subunits.[6] NPTX2 is a pre-synaptic protein that acts on post-synaptic excitatory synapses.[7] Figure adapted from.[8]

Results

Sabirnetug-Associated Changes in CSF Synaptic Biomarkers Neurogranin & VAMP2 Indicate Downstream Pharmacology After 3 Doses

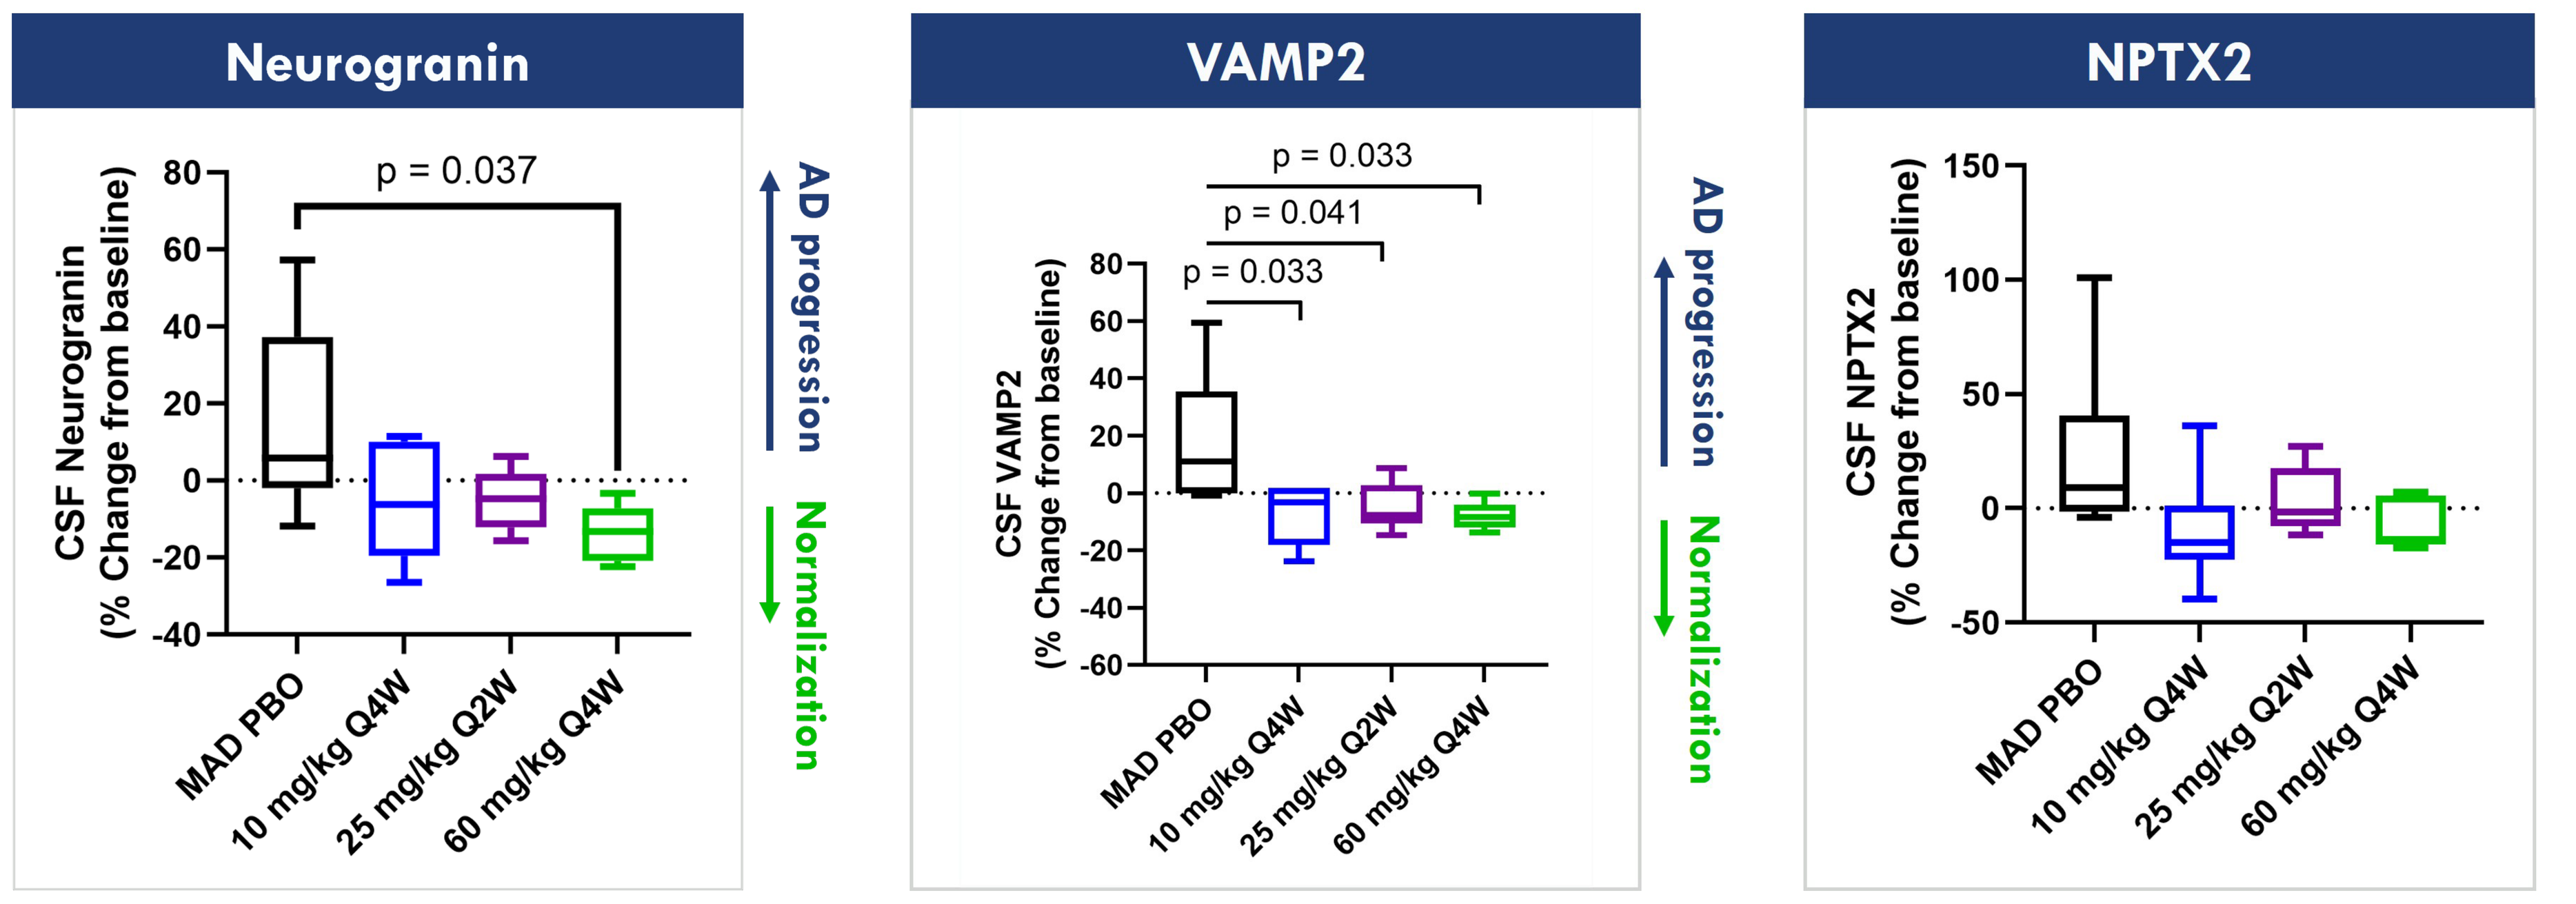

Neurogranin
CSF Neurogranin (% Change from baseline)
p = 0.037
VAMP2
CSF VAMP2 (% Change from baseline)
p = 0.033
p = 0.041
p = 0.033
NPTX2
CSF NPTX2 (% Change from baseline)
MAD PBO
10 mg/kg Q4W
25 mg/kg Q2W
60 mg/kg Q4W
AD progression
Normalization

Figure 4. Synaptic biomarkers measured in CSF in the multiple ascending dose (MAD) cohorts. Significant p-values from unpaired, two-sided Student's t test ($\alpha = 0.05$) are listed above the plots. n = 8 subjects/treated group; 6 subjects in pooled placebo (PBO). Neurogranin was measured via ELISA (EUROIMMUN), VAMP2 via ELISA (ADx prototype), and NPTX2 via ELISA (Fujirebio). All assays were run at Amsterdam UMC. Box and whisker plots show median (line), interquartile range (boxes), and minimum/maximum (whiskers). The direction of AD progression & normalization is indicated to the right of the neurogranin & VAMP2 plots. It is less clear from the literature whether an increase or decrease of CSF NPTX2 levels would represent a therapeutic benefit in the treatment population of INTERCEPT-AD (MCI & mild AD). Studies have observed lower CSF NPTX2 in AD vs. cognitively normal controls[7,9-11] and lower CSF NPTX2 values in MCI vs. cognitively normal controls.[12,13] In contrast, CSF NPTX2 levels increased in MCI patients that are within 2 years of symptom onset.[14] No sabirnetug-dependent trends were observed for any biomarker in the single-ascending dose (SAD) cohorts.

Plasma Biomarkers Trend Towards Normalization 1-2 weeks After Last of 3 Sabirnetug Doses

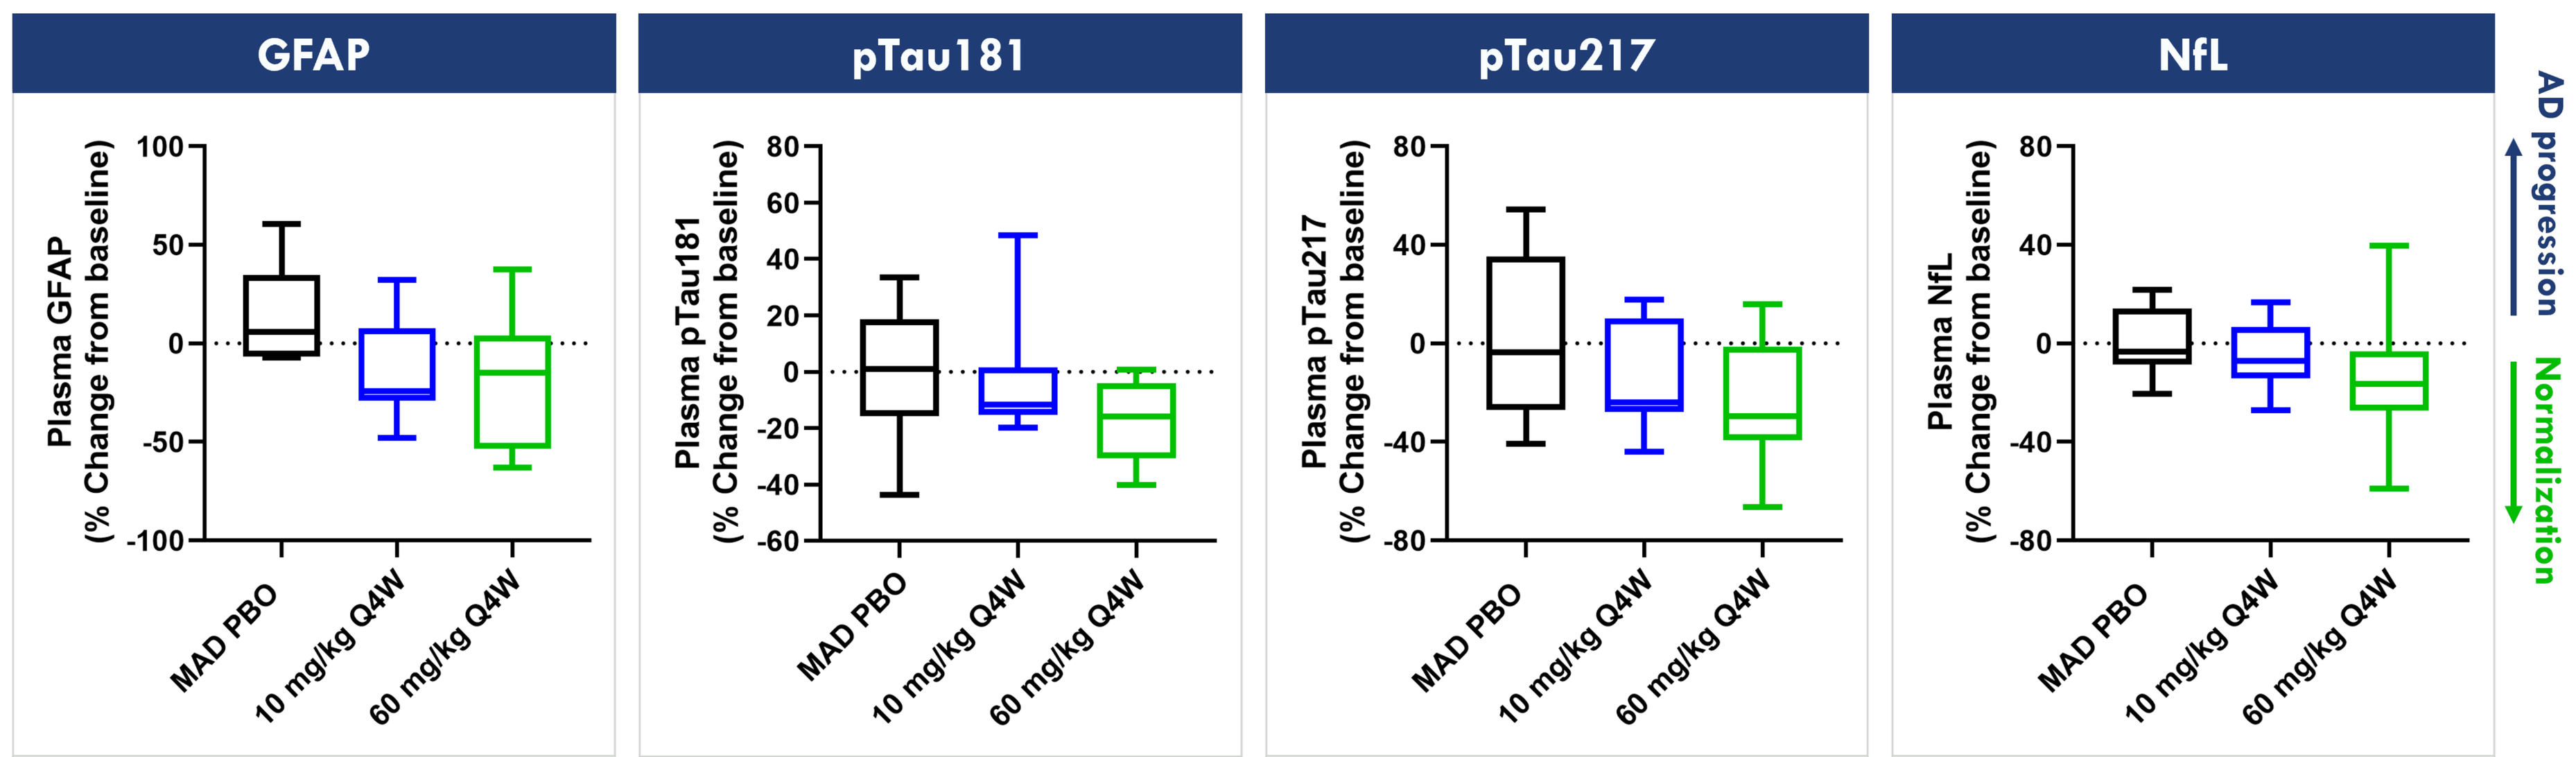

GFAP
Plasma GFAP (% Change from baseline)
pTau181
Plasma pTau181 (% Change from baseline)
pTau217
Plasma pTau217 (% Change from baseline)
NfL
Plasma NfL (% Change from baseline)
MAD PBO
10 mg/kg Q4W
60 mg/kg Q4W
AD progression
Normalization

Sabirnetug Washout is Associated with a Reduction of the Pharmacodynamic Response

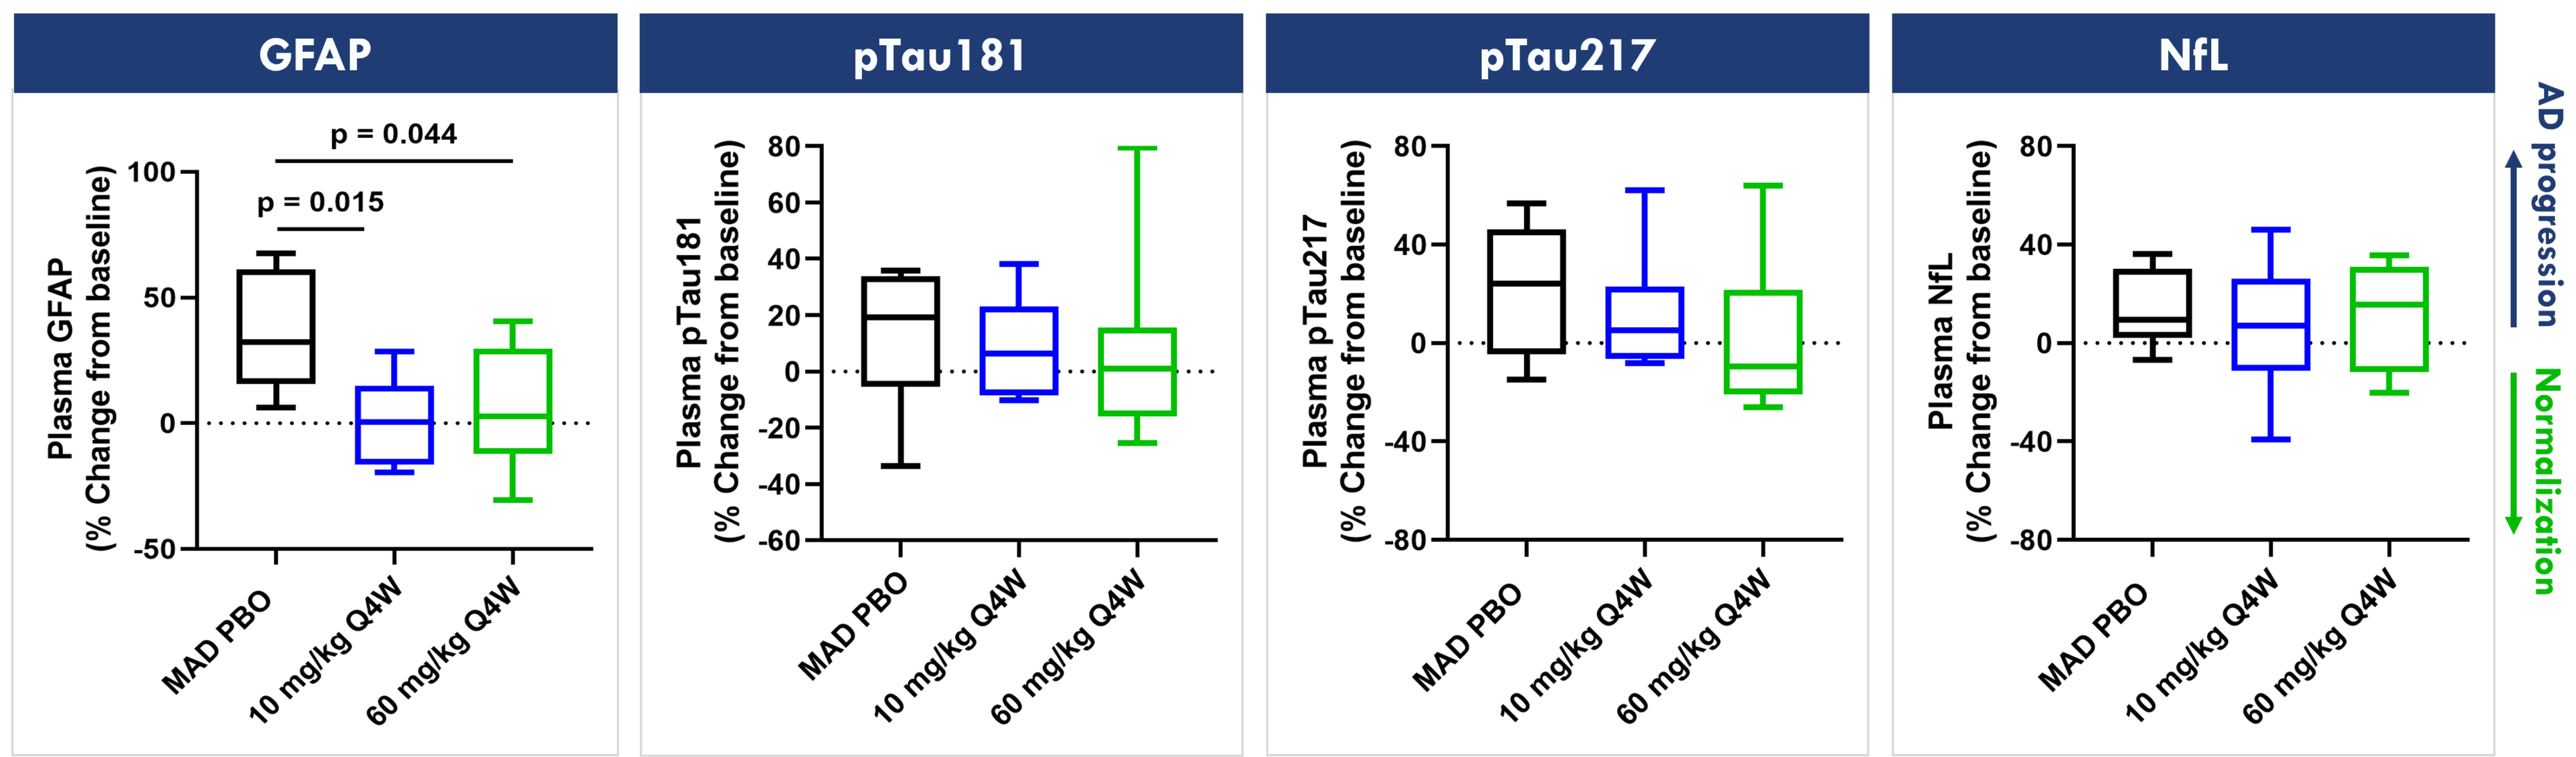

GFAP
Plasma GFAP (% Change from baseline)
p = 0.044
p = 0.015
pTau181
Plasma pTau181 (% Change from baseline)
pTau217
Plasma pTau217 (% Change from baseline)
NfL
Plasma NfL (% Change from baseline)
MAD PBO
10 mg/kg Q4W
60 mg/kg Q4W
AD progression
Normalization

Figure 5. Biomarkers measured in plasma in the MAD Q4W cohorts. Top) 1st post-dose sampling time, 1-2 weeks after last dose. Bottom) 2nd post-dose sampling time, 10-20 weeks after last dose. Significant p-values from unpaired, two-sided Student's t test ($\alpha = 0.05$) are listed above the plots. n = 8 subjects/treated group; 6 subjects in pooled PBO. GFAP and NfL were measured in the SimoA N4PE 4-Plex assay (Quanterix) with Aβ42 & Aβ40. Aβ data are not shown due to sabirnetug drug interference in the assays. No trends were observed in cohort 7 (25 mg/kg Q2W) or the SAD cohorts. Data plotted without cohort 7 to facilitate visualization of trends in Q4W cohorts. pTau181 & pTau217 were measured by single-plex SimoA assays (Quanterix & AlzPath/Quanterix, respectively). All assays were run at Amsterdam UMC. The direction of AD progression & normalization is indicated to the right of the plots.

RESEARCH HIGHLIGHTS

- Three administrations of sabirnetug lowered CSF levels of both pre- & post-synaptic proteins, consistent with its proposed mechanism of action to inhibit Aβ oligomer synaptic binding.
- VAMP2 appears most sensitive to sabirnetug in this study, lowering significantly in all 3 MAD cohorts after sabirnetug treatment.
- Plasma biomarkers trended towards normalization with sabirnetug treatment but the trends did not reach statistical significance.
- Biomarker responses to longer-term sabirnetug treatment & their relationship to clinical outcomes will be evaluated in the ongoing 18-month ALTITUDE-AD phase 2 study (NCT06335173).

1. Cline et al. J Alzheimers Dis, 2018;61(s1):S567-S610. 2. Krafft et al. Front Neurosci, 2022:16:848215. 3. Data on file. 4. Hampel et al. Nat Rev Neurol, 2021;17(9):580-589. 5. O'Day. Int J Mol Sci. 2020;21(19):7344. 6. Goossens et al. Alzheimers Res Ther 2023;15:186. 7. Xiao et al. Elife 2017;6:e23798. 8. Das et al. Alzheimers Res Ther, 2023;15:62. 9. Galasko et al. Alzheimers Dement, 2019;5:871-882. 10. Sathe et al. Proteomics Clin Appl, 2019;13(4):e1800105. 11. Nilsson et al. Brain, 2024;awae032. 12. Libiger et al. Alzheimers Dement, 2021;17(12):1976-1987. 13. Soldan et al. Front Aging Neurosci, 2019;11:132. 14. Massa et al. J Neurol, 2024;271(4):1999-2009.